Abstract number: 581

Preliminary Phase 1 results of clinical trial investigating BI-1910, a Tumor Necrosis Factor Receptor 2 (TNFR2) agonist in solid cancer tumor patients

Authors: T. Hernandez Guerrero[A], B. Doger de Speville[B], J. Yachnin[C], K. Staal Rohrberg[D], I. Moreno Candilejo[E] A. Carneiro[F], A. Falcon Gonzalez[G], P. Holmkvist[H], I. Karlsson[H], D. Lindahl[H], M. Meller[H], L. Mårtensson[H], O. Schönborn-Kellenberger[I], A. Sundstedt[H], D. Piliuhin[H], M. Vaapil[H], J. Wallin[H], I. Teige[H], B. Frendeus[H,J], A. McAllister[H]

[A] START Barcelona – HM Nou Delfos Hospital, Barcelona, Spain; [B] START Madrid Hospital Fundacion Jimenez Diaz, Madrid, Spain; [C] Karolinska Comprehensive Cancer Center, Karolinska Universitetssjukhuset, Solna, Sweden; [D] Department of Oncology, Copenhagen University Hospital - Rigshospitalet, Copenhagen, Denmark; [E] START Madrid - CIOCC - HM Sanchinarro, Madrid, Spain; [F] Skånes Universitetssjukhus – Lund, Sweden; [G] Hospital Universitario Virgen del Rocío, Sevilla, Spain; [H] BioInvent International AB, Lund, Sweden; [I] Cogitars GmbH, Heidelberg, Germany; [J] Centre for Cancer Immunology, University of Southampton, Southampton, United Kingdom

Rationale for Developing BI-1910

TNFR2 is a novel promising target for cancer immune therapy, and TNFR2 has been proposed as a costimulatory receptor for T-cell activation. Early clinical development of ligand-blocking mAbs targeting TNFR2, such as BI-1808, indicate that modulating the pathway may lead to clinical responses with favourable tolerability.

We have developed BI-1910, a TNFR2 targeting agonist. BI-1910 does not block the interaction between TNFR2 and its ligand TNF-α. Non-clinical in vivo data demonstrate enhancement of functional activity of TNFR2-expressing lymphocytes, including CD8+ and CD4+ T cells, as well as NK cells, resulting in tumor regression and ultimately immune rejection.

We report here a Phase 1 clinical trial of the TNFR-2 targeting antibody BI-1910 in subjects with advanced/metastatic solid tumors.

Mechanism of action of BI-1910

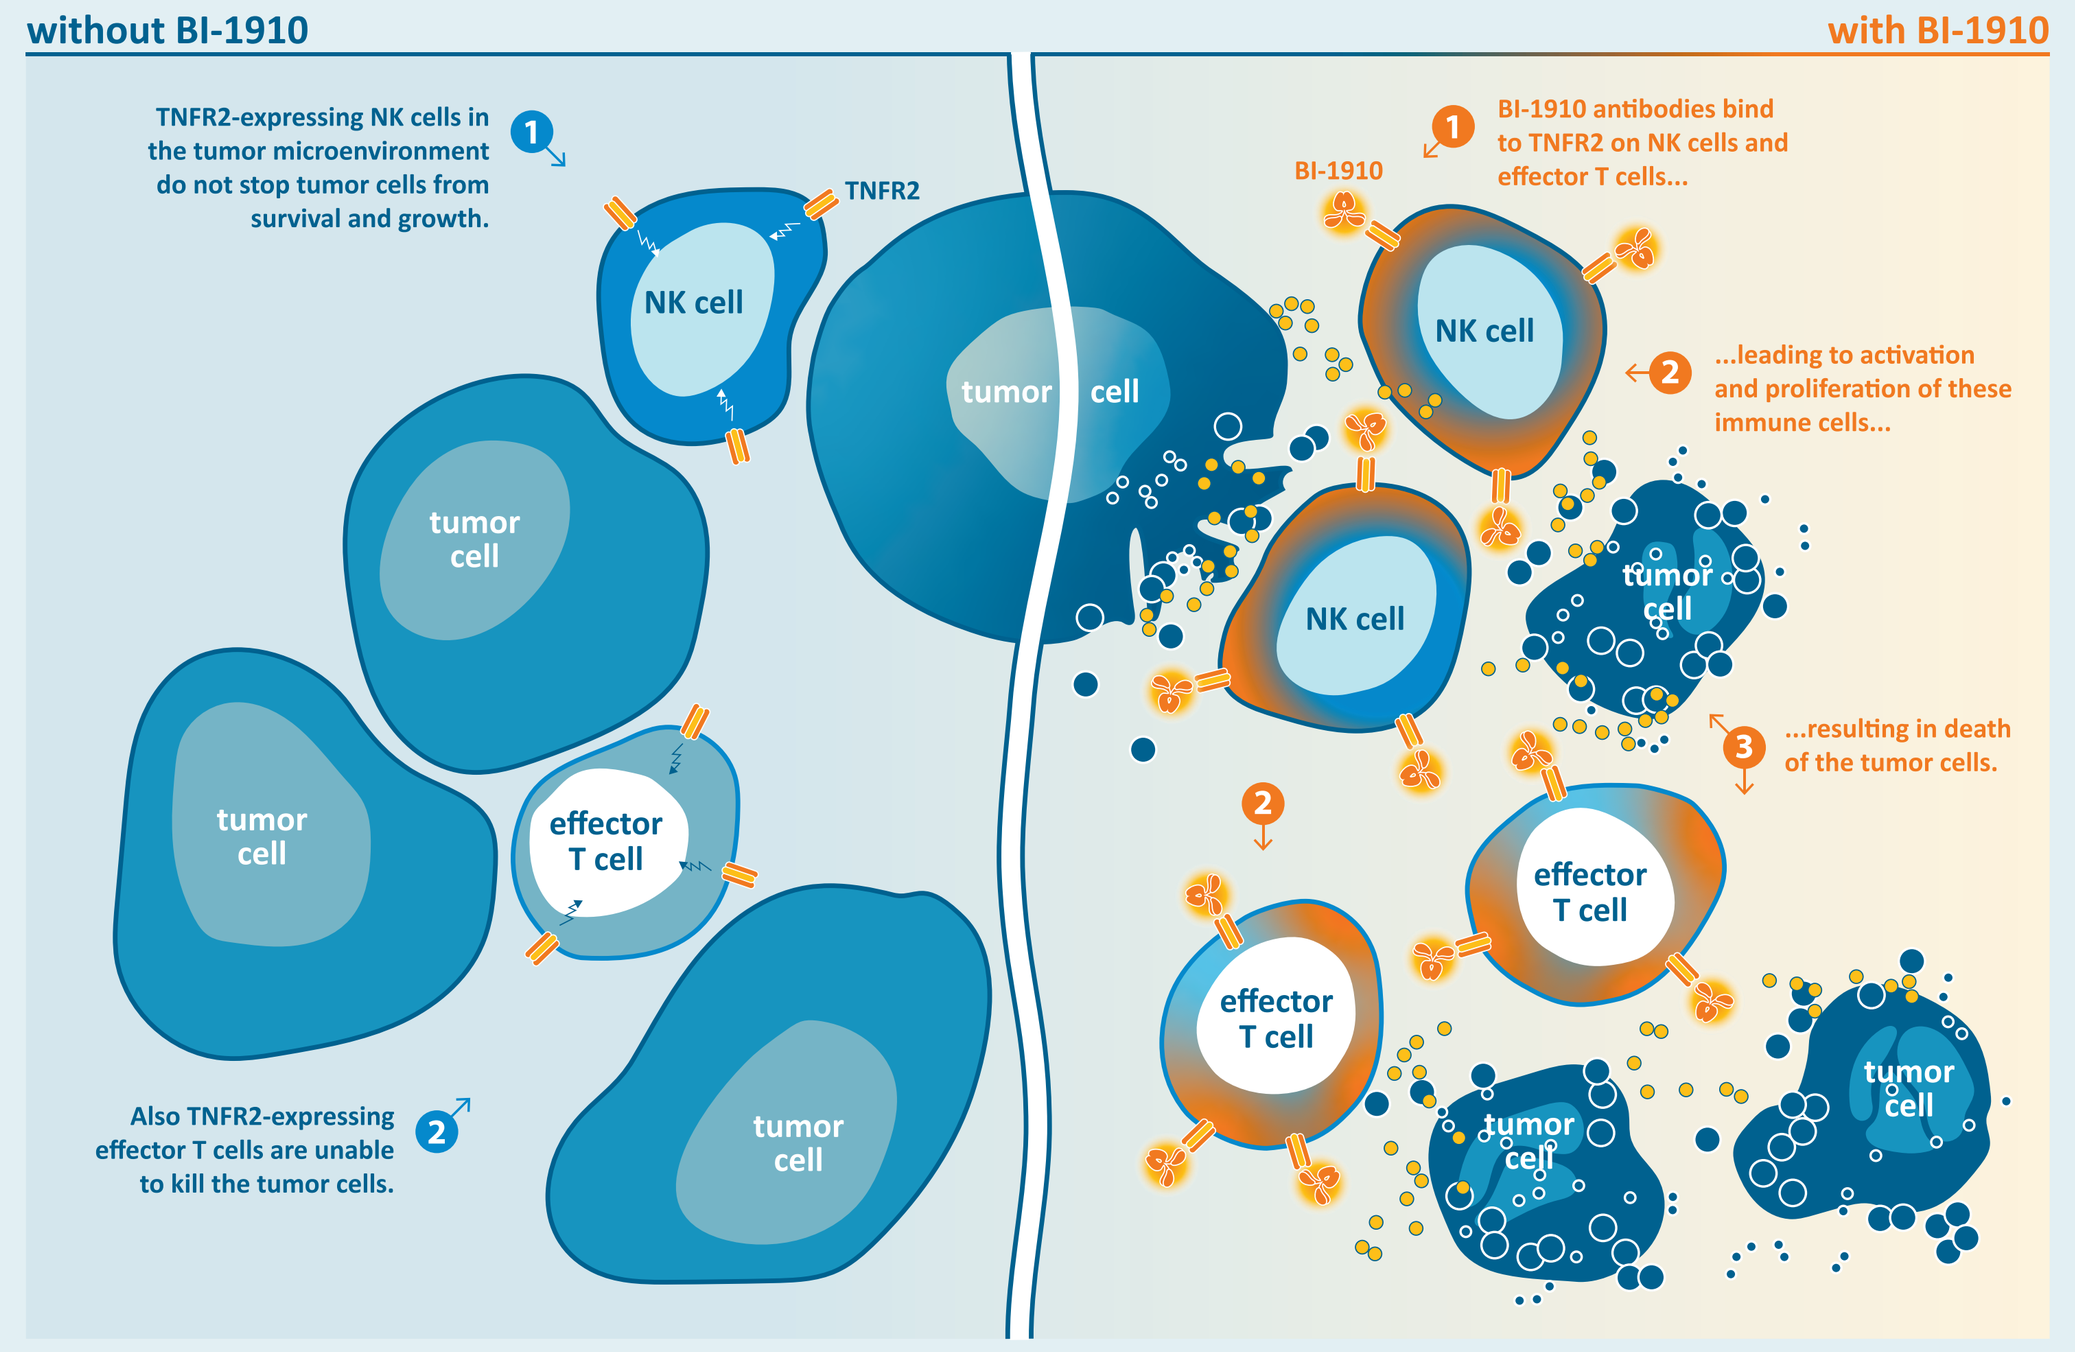
without BI-1910
with BI-1910
TNFR2-expressing NK cells in the tumor microenvironment do not stop tumor cells from survival and growth.
TNFR2
NK cell
tumor cell
effector T cell
Also TNFR2-expressing effector T cells are unable to kill the tumor cells.
BI-1910
BI-1910 antibodies bind to TNFR2 on NK cells and effector T cells...
...leading to activation and proliferation of these immune cells...
...resulting in death of the tumor cells.

About TNFR2

- High expression on T regs, activated/ memory CD8+ T cells, NK cells, also DC's and several myeloid cells, e.g. monocytes
 - – Shares ligands with TNFR1
 - – Membrane bound TNF-α binds and signals through TNFR2
- TNFR2 has been shown to be critical for T reg proliferation and survival
- Proposed as a co-stimulatory factor for T cells proliferation and activation (similar to e.g. OX40 or 4-1BB)

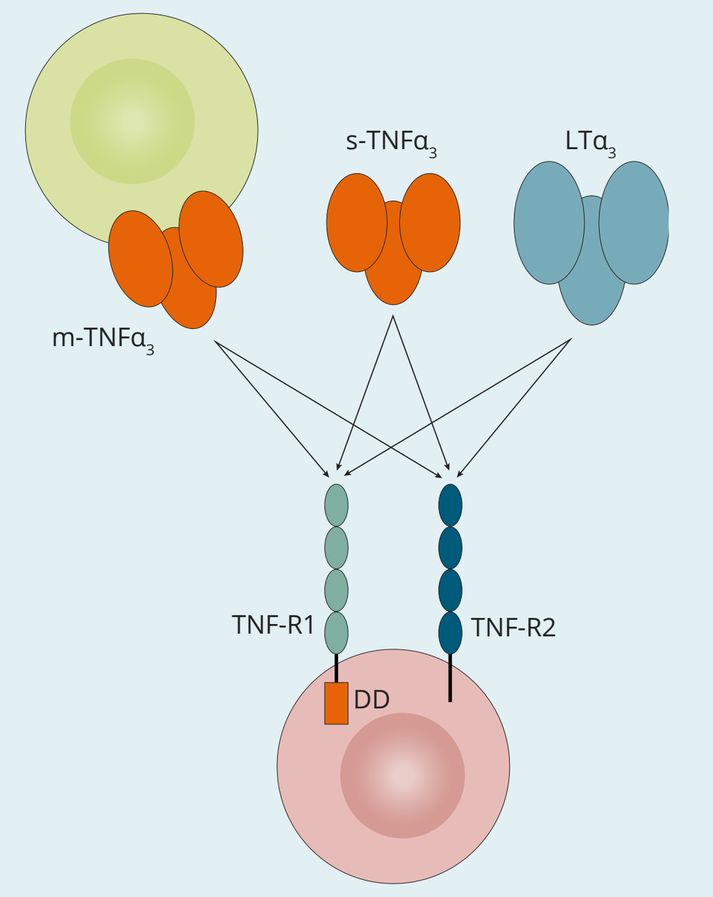
m-TNFα3
s-TNFα3
LTα3
TNF-R1
TNF-R2
DD

BI-1910 exhibits an excellent safety profile, combined with strong evidence CD8+ T-cell activation associated with disease control in heavily pretreated patients

Strong T-cell activation in patients with disease control

CD4+ memory T-cell expansion was observed in all patients, with higher increases in patients with disease control. This effect was strongest after first administered dose (Fig. B).

Stronger CD4+ T-cell activation, as measured by 4-1BB and PD1 upregulation was observed in patients with long-term (over 6 months) stable disease, indicating that immune cell activation underlain the disease stabilization.

CD8+ memory T-cell expansion was observed in all patients, and in contrast to CD4+ T cells, this continued to increase with repeat dosing (Fig. A). Prolonged disease control >6 months correlated with higher levels of post-treatment CD8+ T-cell expansion.

Similarly, over time an increase in the activation markers 4-1BB and PD1 was observed on CD8+ T cells in patients with long-term stable disease. Following the third dose (day 43), there was a clear separation between patients experiencing long-term disease control and others, again strongly pointing to T-cell activation as a key component behind the observed disease stabilization.

Patients with lasting disease control also expressed a higher increase in soluble TNFR2 compared to patients with shorter or no disease control, suggesting more pronounced agonism in these patients (Fig. C).

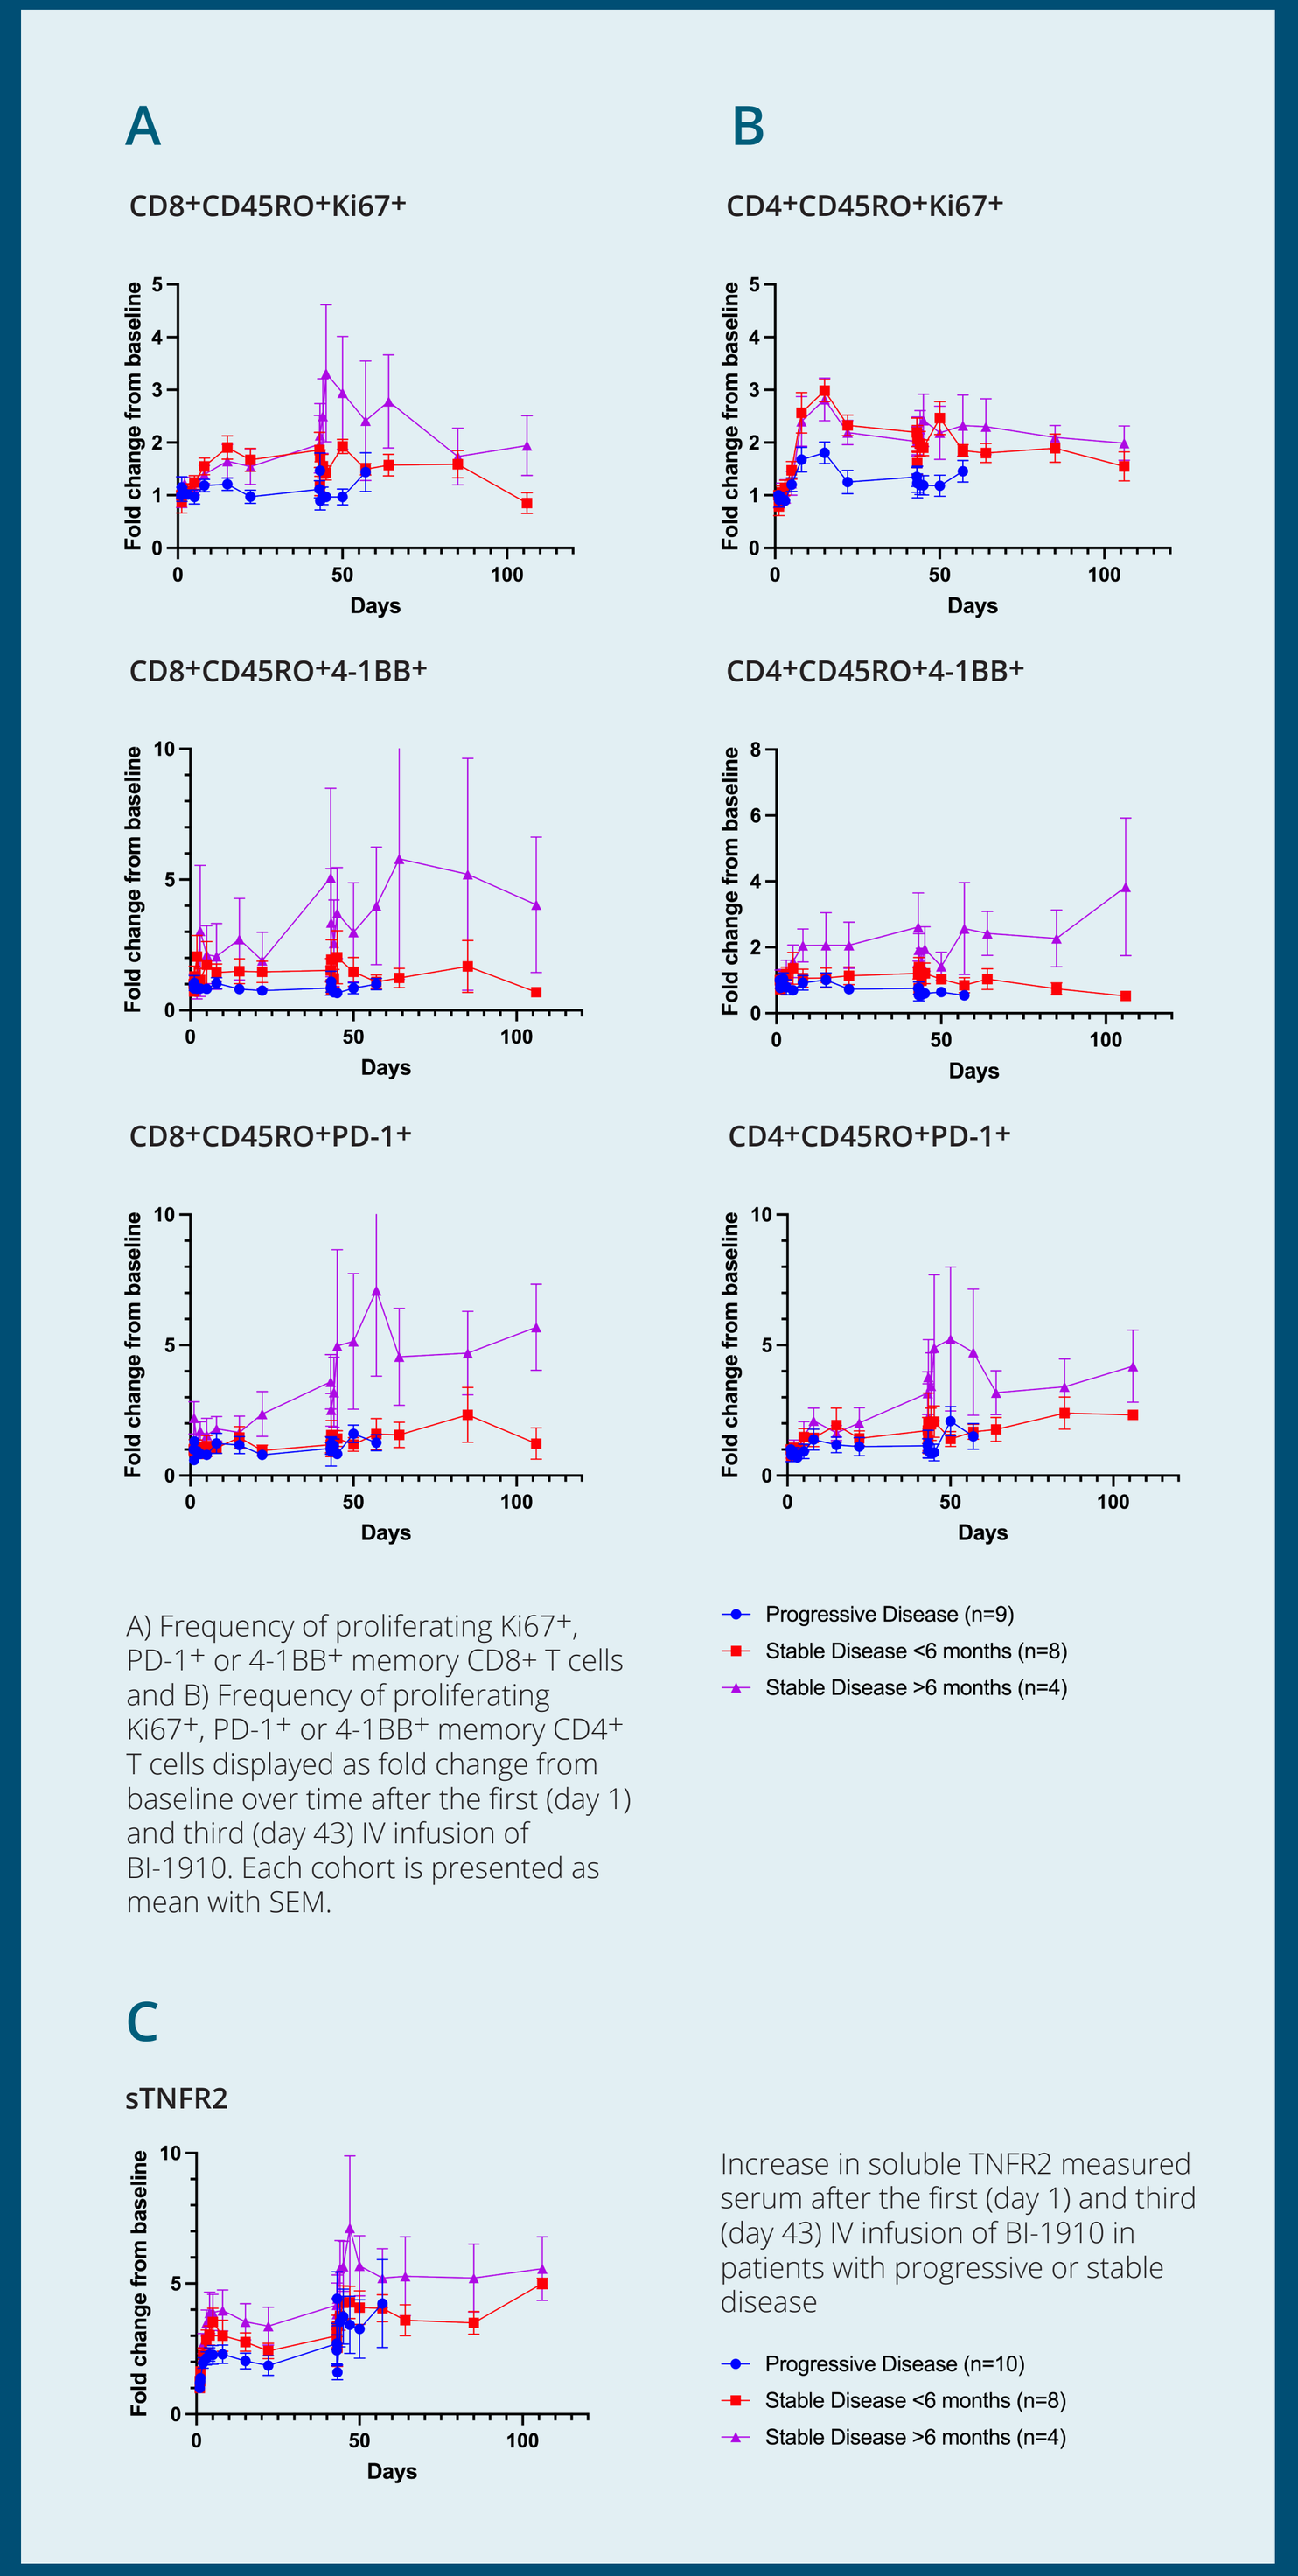
A
B
CD8+CD45RO+Ki67+
CD4+CD45RO+Ki67+
CD8+CD45RO+4-1BB+
CD4+CD45RO+4-1BB+
CD8+CD45RO+PD-1+
CD4+CD45RO+PD-1+
Fold change from baseline
Days
Progressive Disease (n=9)
Stable Disease <6 months (n=8)
Stable Disease >6 months (n=4)
C
sTNFR2
Progressive Disease (n=10)
Stable Disease <6 months (n=8)
Stable Disease >6 months (n=4)

A) Frequency of proliferating Ki67+, PD-1+ or 4-1BB+ memory CD8+ T cells and B) Frequency of proliferating Ki67+, PD-1+ or 4-1BB+ memory CD4+ T cells displayed as fold change from baseline over time after the first (day 1) and third (day 43) IV infusion of BI-1910. Each cohort is presented as mean with SEM.

Increase in soluble TNFR2 measured serum after the first (day 1) and third (day 43) IV infusion of BI-1910 in patients with progressive or stable disease

Case study: Ovarian cancer

Biopsies from an ovarian cancer patient taken at pre-treatment and 10 weeks after first dose of BI-1910.

Sections were stained for CD3, CD4, CD8, FOXP3 and panCytokeratin (Ultivue) and the density of key phenotypes (T regulatory cells and cytotoxic T cells) were analyzed in the entire tumor area using Visiopharm software.

Images showing T regulatory cells (FOXP3, pink), cytotoxic T cells (CD8, green), and tumor cells (panCK, cyan) at pre-dose and week 10. The patient has ongoing stable disease 1 year after start of treatment.

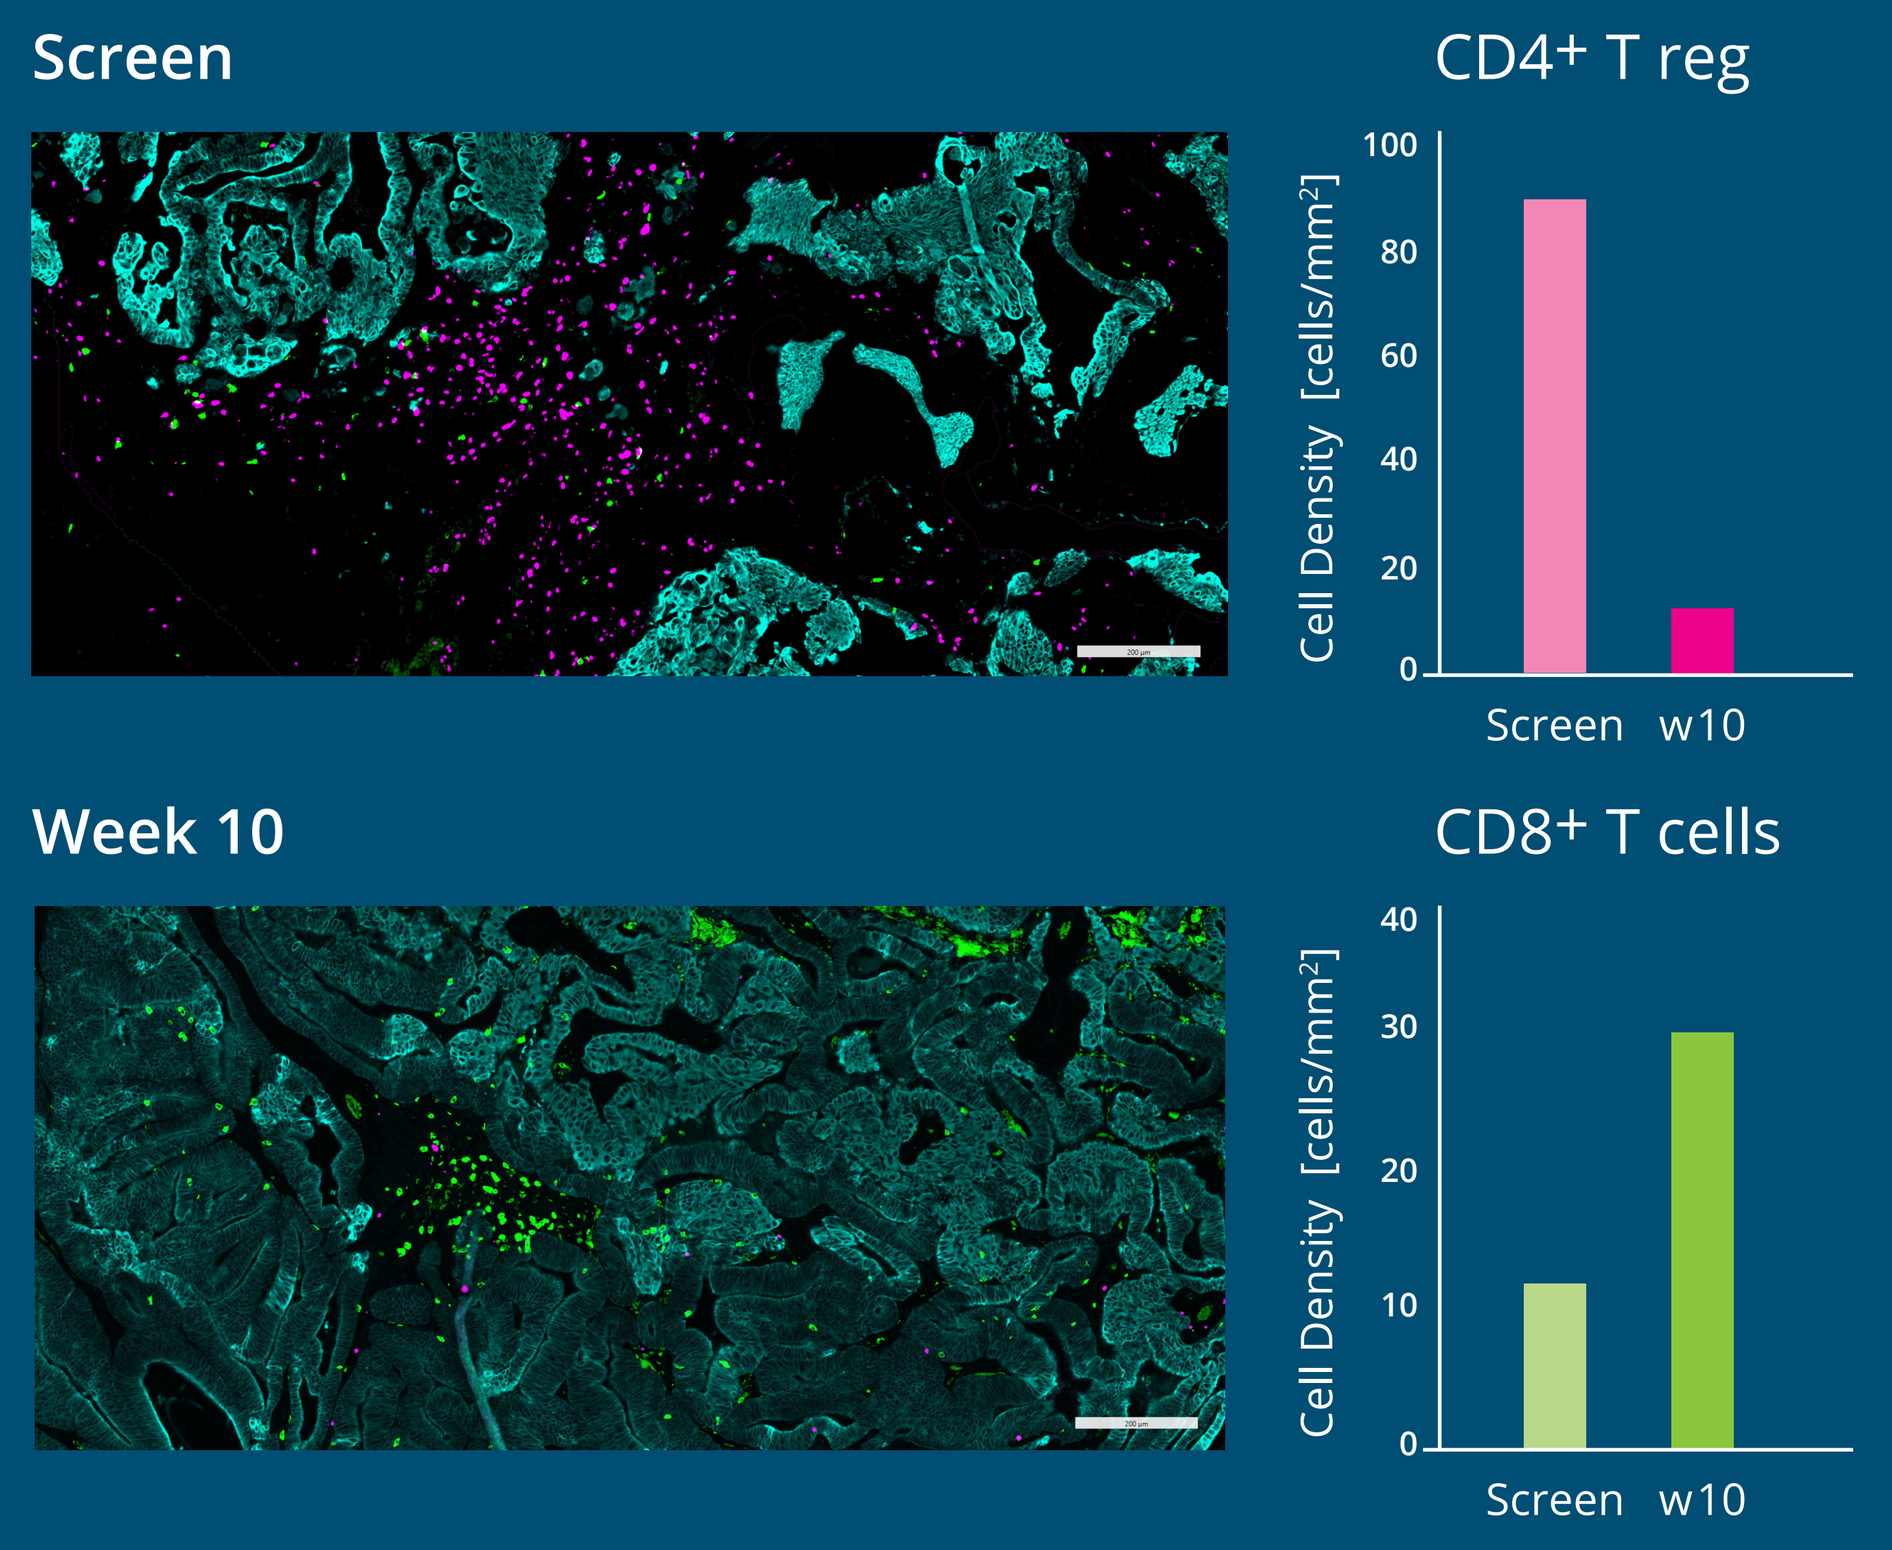
Screen
CD4+ T reg
Week 10
CD8+ T cells
Cell Density [cells/mm²]
Screen
w10

Background

Tumor necrosis factor (TNF) is a pro-inflammatory cytokine that induces inflammatory responses and cell death. TNF interacts with two receptors: TNFR1, expressed across various cell types, and TNFR2, mainly expressed on cells of the immune system. Macrophages and regulatory T cells constitutively express TNFR2. In effector T cells, it increases after receptor stimulation.

TNFR2 is associated with both pro-inflammatory and immunoregulatory functions, is vital for Treg survival, and acts as a potent costimulatory molecule on CD8+ T cells. It promotes inflammation and has emerged as a promising target for cancer immunotherapy.

BI-1910 is an agonistic human IgG2 mAb targeting TNFR2. BI-1910 stimulates T cells and enhances the activation of both CD4+ and CD8+ T cells. It binds selectively to human TNFR2 without inhibiting TNF-α binding. In preclinical models, BI-1910 combined with anti-PD-1 showed additive anti-tumor activity, justifying clinical evaluation with pembrolizumab.

Study overview

The study enrolled subjects with advanced/metastatic solid tumors who had progressed after standard therapy.

Objectives included assessment of safety/tolerability, pharmacokinetics, pharmacodynamics and efficacy of BI-1910 as single agent.

Dose escalation was performed using a BLRM design, exploring dose levels from 4 mg through 900 mg Q3W.

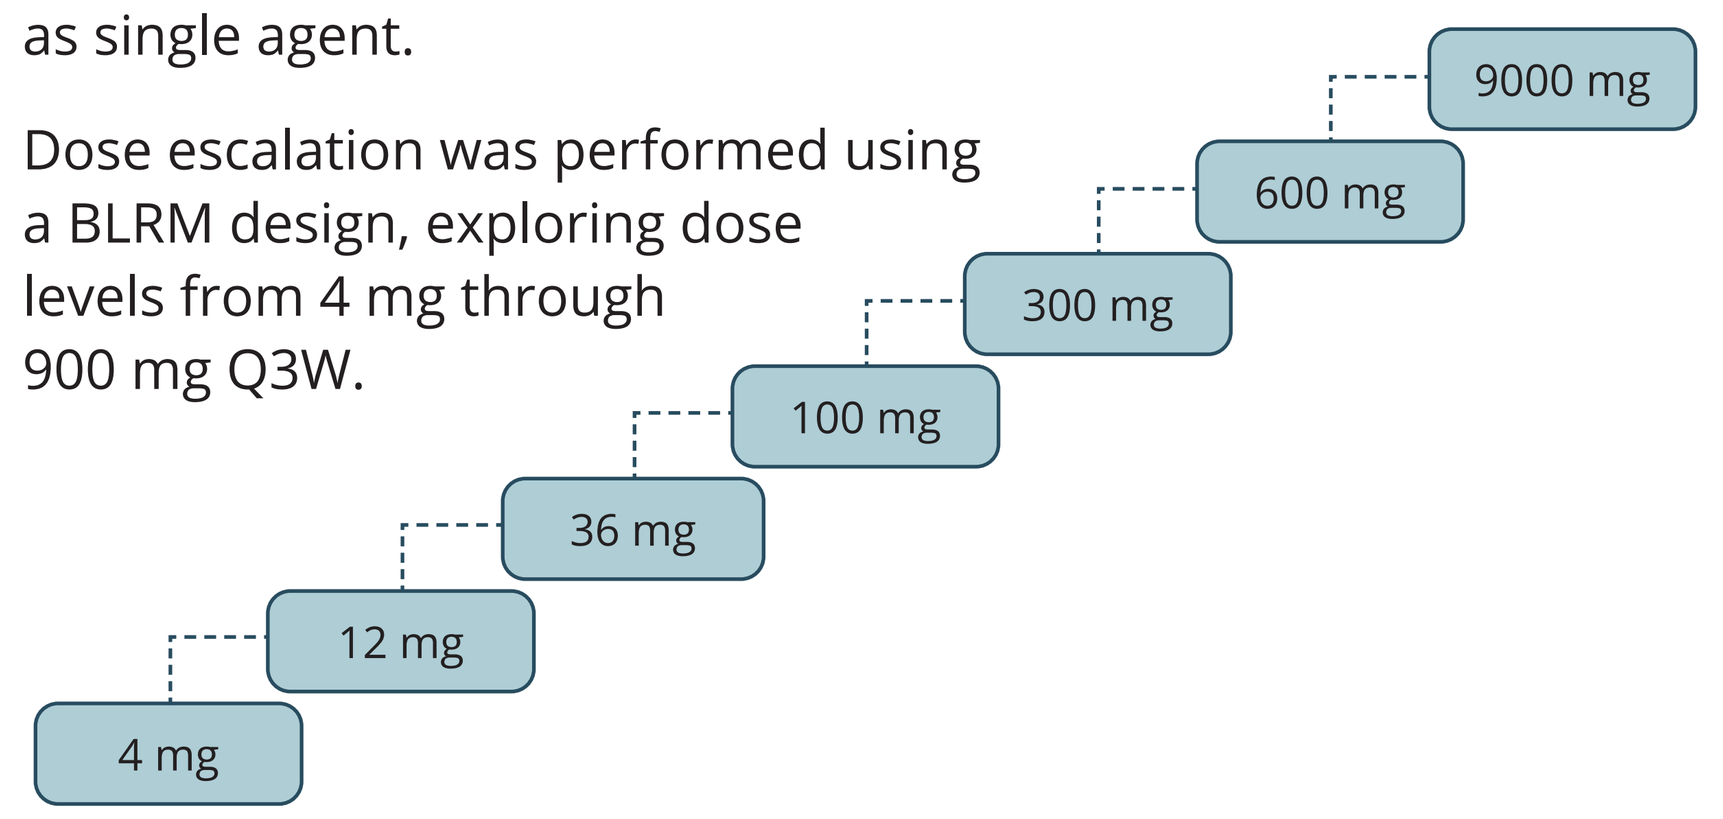
9000 mg
600 mg
300 mg
100 mg
36 mg
12 mg
4 mg

Results

- As of September 6th, 2025, 26 subjects received doses of 4 to 900 mg BI-1910 as a single agent Q3W. Doses ≥300 mg Q3W resulted in full receptor occupancy.
- Treatment is overall well tolerated, with no DLT:s observed. The most common AE was fatigue.
- The best clinical response was stable disease in 12 patients, of whom five with neuroendocrine, salivary gland, endometrial, and ovarian tumors are currently exhibiting disease control for more than 6 months.
- T-cell activation and proliferation related to treatment (Fig. A-C) was observed in patients with stable disease.

Overview of most common TEAE:s related to BI-1910

BI-1910 single agent	Administration route: IV							
Dose	4 mg	12 mg	36 mg	100 mg	300 mg	600 mg	900 mg	Total
Number of subjects	*1*	*2*	*1*	*3*	*8*	*7*	*4*	*26*
Subjects with any grade Fatigue/ Asthenia	0	0	1 (100%)	0	2 (25%)	3 (43%)	1 (25%)	7 (27%)
Subjects with ≥Grade 3 Fatigue/ Asthenia	0	0	0	0	1 (13%)	0	0	1 (4%)
Subjects with any grade Anaemia	0	0	0	0	2 (25%)	0	0	2 (8%)
Subjects with ≥Grade 3 Anaemia	0	0	0	0	0	0	0	0
Subjects with any grade Dry eye	0	0	0	0	1 (13%)	1 (14%)	1 (25%)	3 (11%)
Subjects with ≥Grade 3 Dry eye	0	0	0	0	0	0	0	0
Subjects with any grade Hyper/Hypothyreoidsm	0	0	0	0	0	0	1 (25%)	1 (4%)
Subjects with ≥Grade 3 Hyper/Hypothyreoidsm	0	0	0	0	0	0	0	0

SLD change from baseline

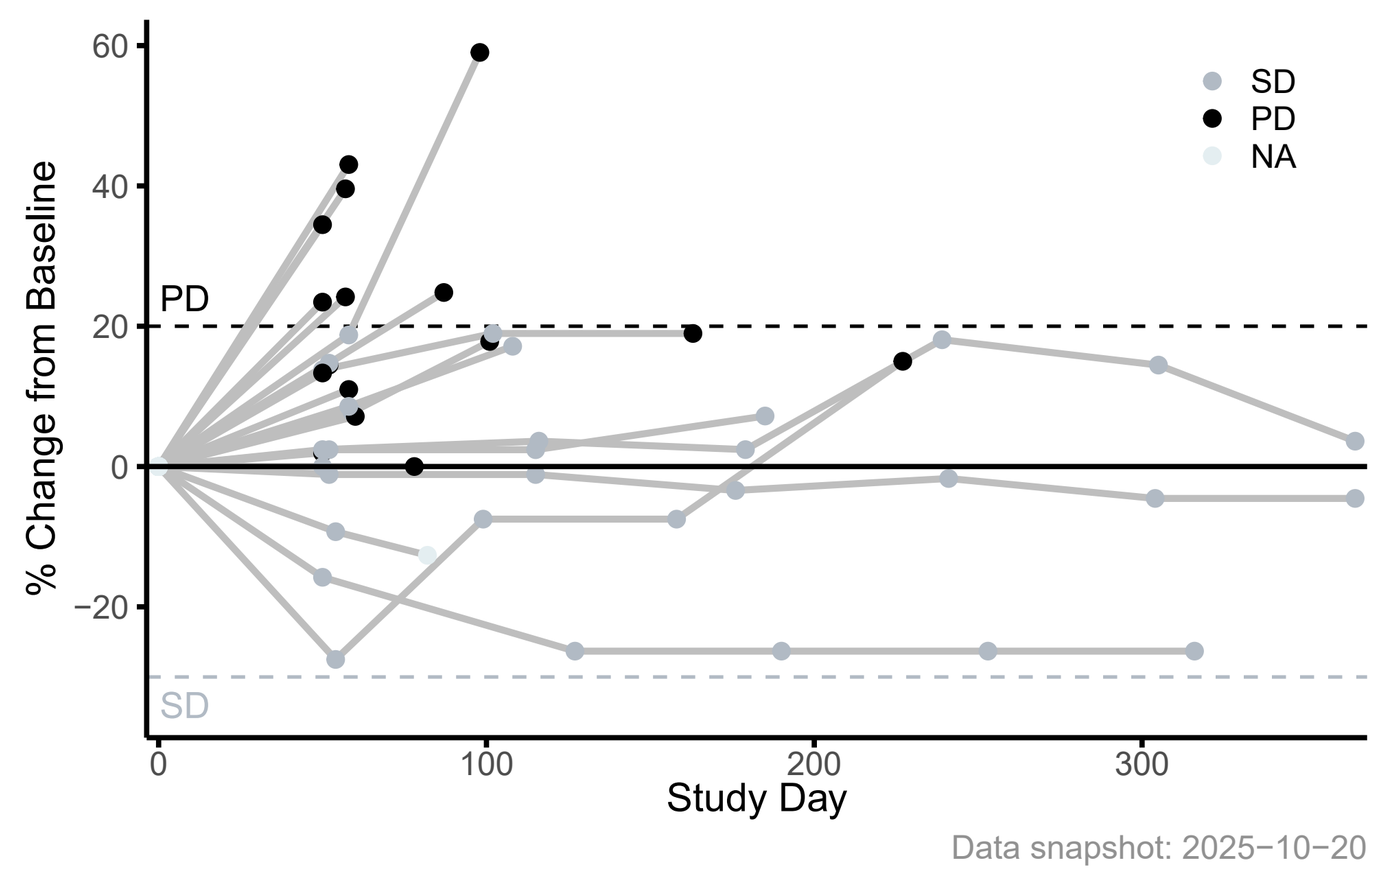
% Change from Baseline
Study Day
PD
SD
SD
PD
NA
Data snapshot: 2025−10−20

Best overall response (RECIST)

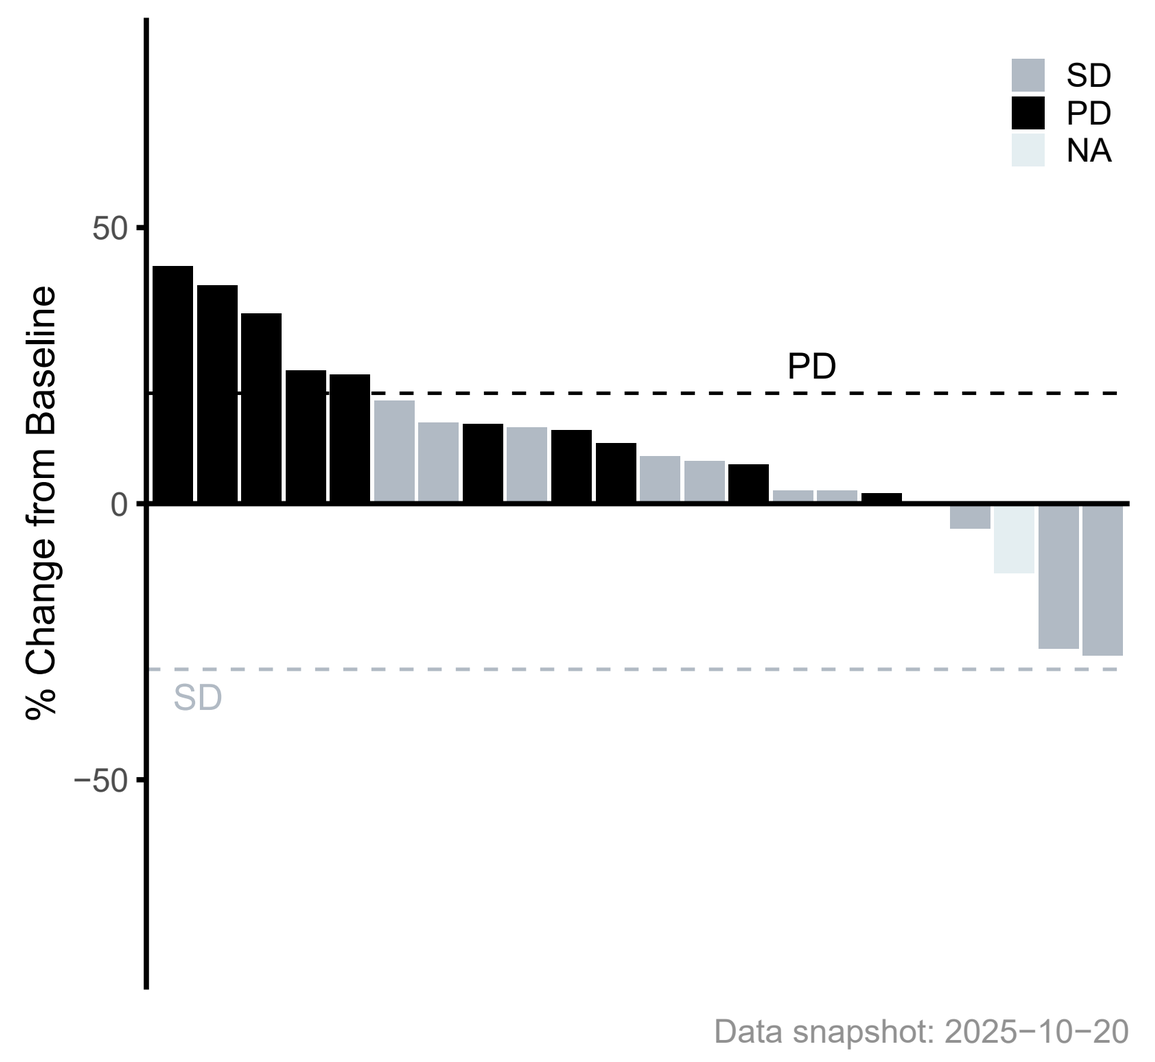
% Change from Baseline
PD
SD
SD
PD
NA
Data snapshot: 2025−10−20

Swimmer plot

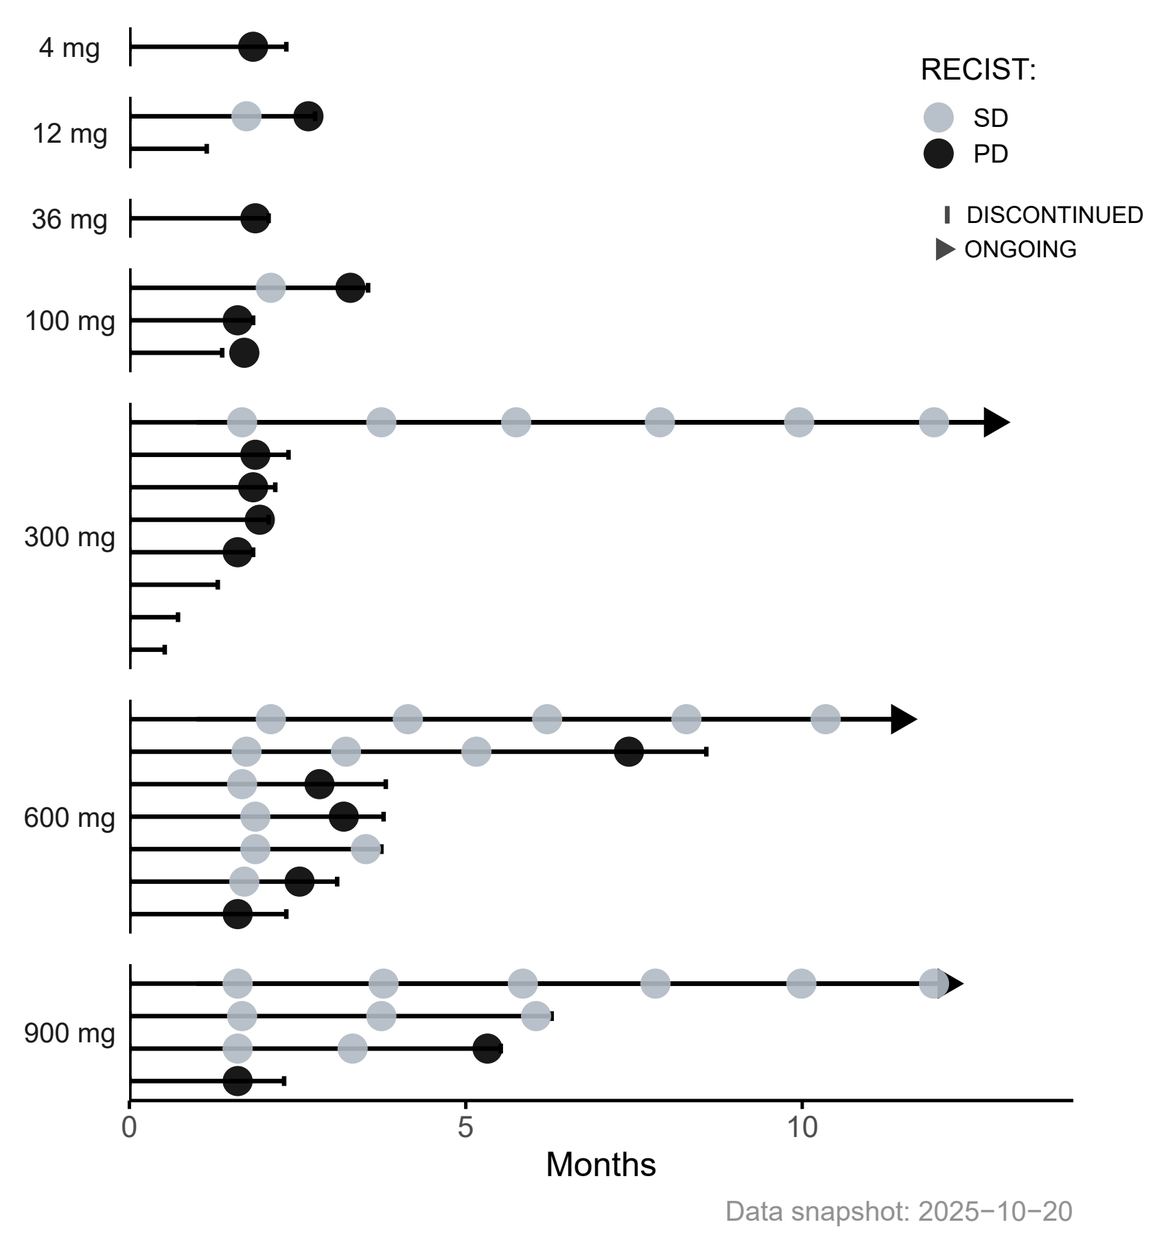
4 mg
12 mg
36 mg
100 mg
300 mg
600 mg
900 mg
Months
RECIST:
SD
PD
DISCONTINUED
ONGOING
Data snapshot: 2025−10−20

Contact information:
clinicaltrials@bioinvent.com

Study identifier:
ClinicalTrials.gov ID: NCT06205706

Acknowledgements
We extend our deep appreciation to study participants, their families and clinical research staff.